AMYPAD
Amyloid imaging to prevent Alzheimer´s Disease

Performance of supratentorial white matter reference regions for longitudinal quantification of [^{18}F]Flutemetamol PET scans

Salvadó G[1], Foley C[2], Grecchi E[3], Cardoso MJ[4], Lopes-Alves I[5], Markiewicz P[4], Falcon C[1], Battle M[2],Lammertsma AA[5], Schmidt ME[6], Barkhof F[4,5], Molinuevo JL[1], Gispert JD[1], *on behalf of the AMYPAD Consortium*

1) Barcelonaβeta Brain Research Center (Spain), 2) GE Healthcare (United Kingdom), 3) IXICO (United Kingdom), 4) University College London (United Kingdom), 5) VU Medical Center (The Netherlands), 6) Janssen Pharmaceutica (Belgium)

Background

Multiple studies have shown that the selection of the reference region (RR) on amyloid (Aβ) PET quantification is critical. However, there is no universally optimal RR as it depends on study design and the radiotracer.

Aim of study

To compare the performance of different supratentorial white matter (sWM) reference regions with those of the Centiloid standard [1] for quantification of longitudinal [^{18}F]Flutemetamol PET scans.

Methods

Sample: 125 subjects from the AIBL study [2] with longitudinal MRI and [^{18}F]Flutemetamol PET images (577±55 days apart).

Preprocessing: Centiloid standard method was used to determine SUVr values in the target cortical VOI, with two pipelines:

- **Cross-sectional:** applying it to both time points independently
- **Longitudinal:** applying it to averaged image pairs

Reference regions:

- **Atlas-based** using different atlases and erosions levels
- **Standard Centiloid RRs**
- **Subject-specific** using SPM WM segmentation with successive erosions

Figures of merit:

- **Effect size** of longitudinal change (Cohen's d) tested in individuals with intermediate Centiloid values ([GZ] 15≤CL<30)
- **Plausibility** defined as the percentage of all subjects with non-negative Aβ changes
- **SUVr mean percentage change per year** in stable participants, defined as either positives (CL≥30), or negatives (CL<15) with changes below 0.04 in SUVr [3]

Results

■ Best result ■ Statistically equivalent to the best result ■ Significantly worse than the best result

Cross-sectional pipeline

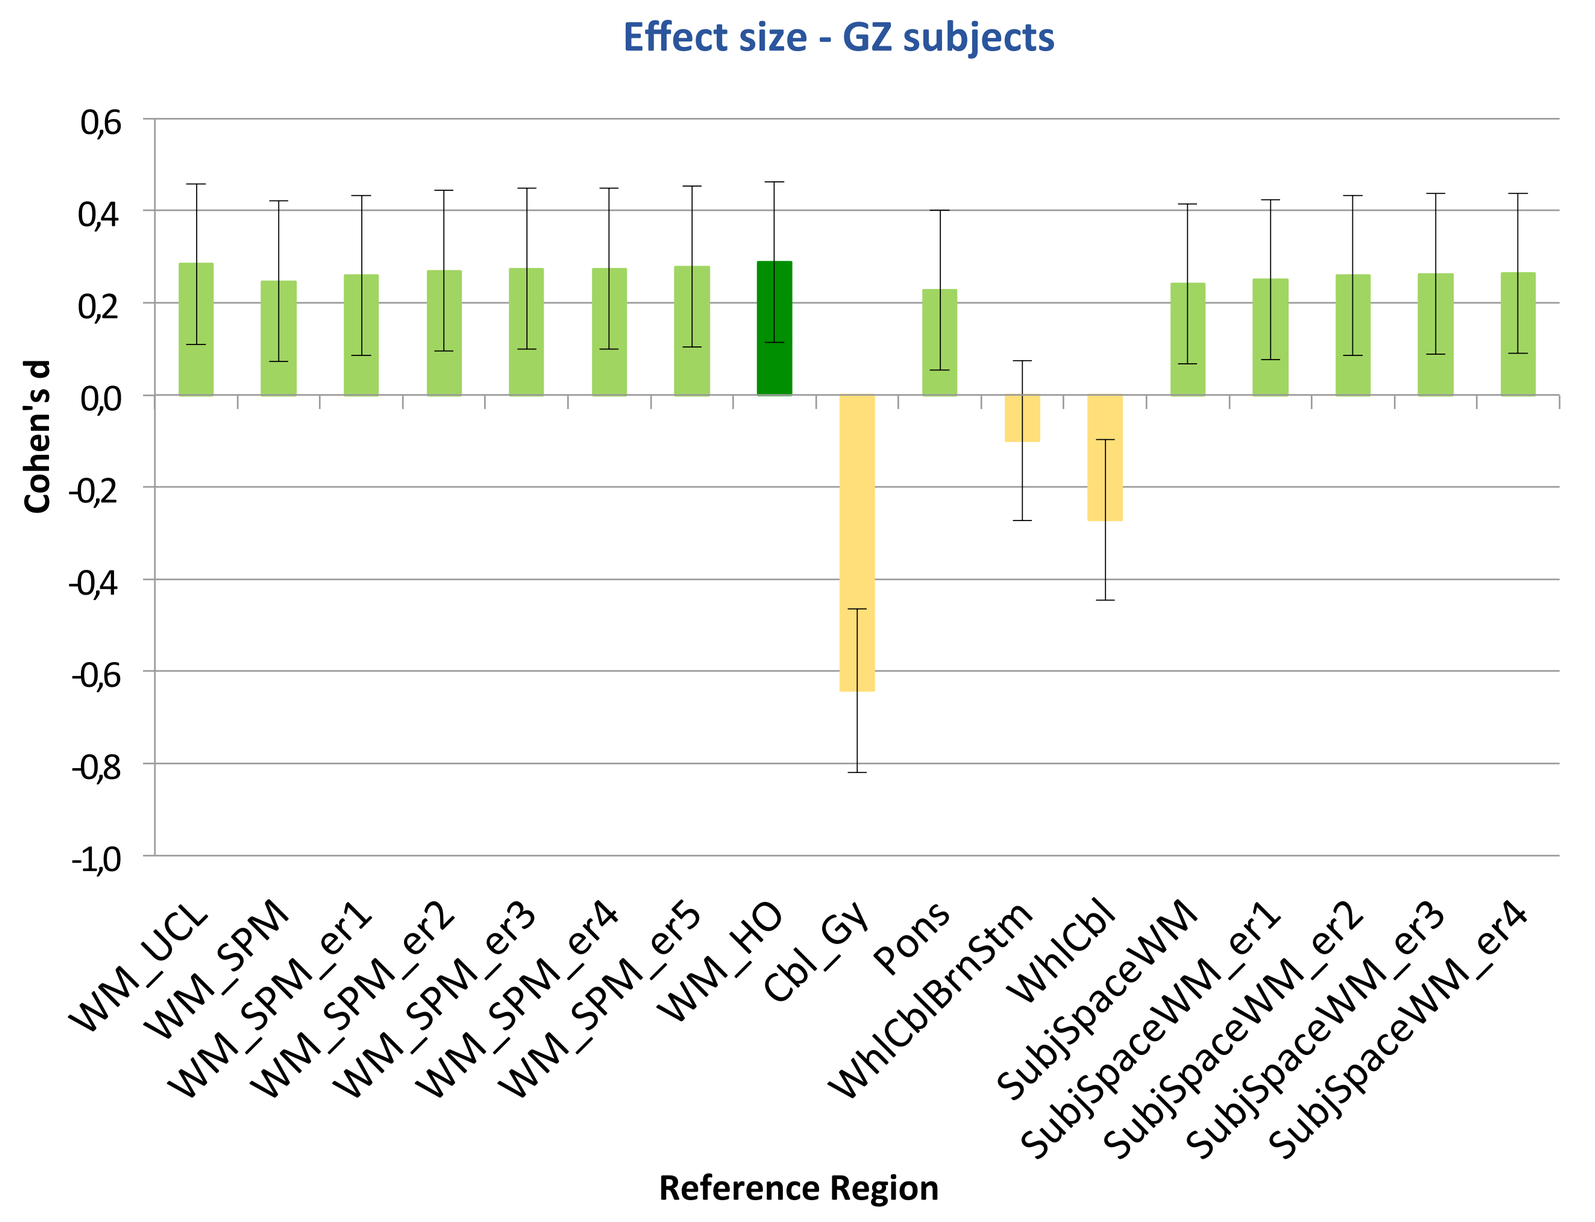
Effect size - GZ subjects
Cohen's d
Reference Region

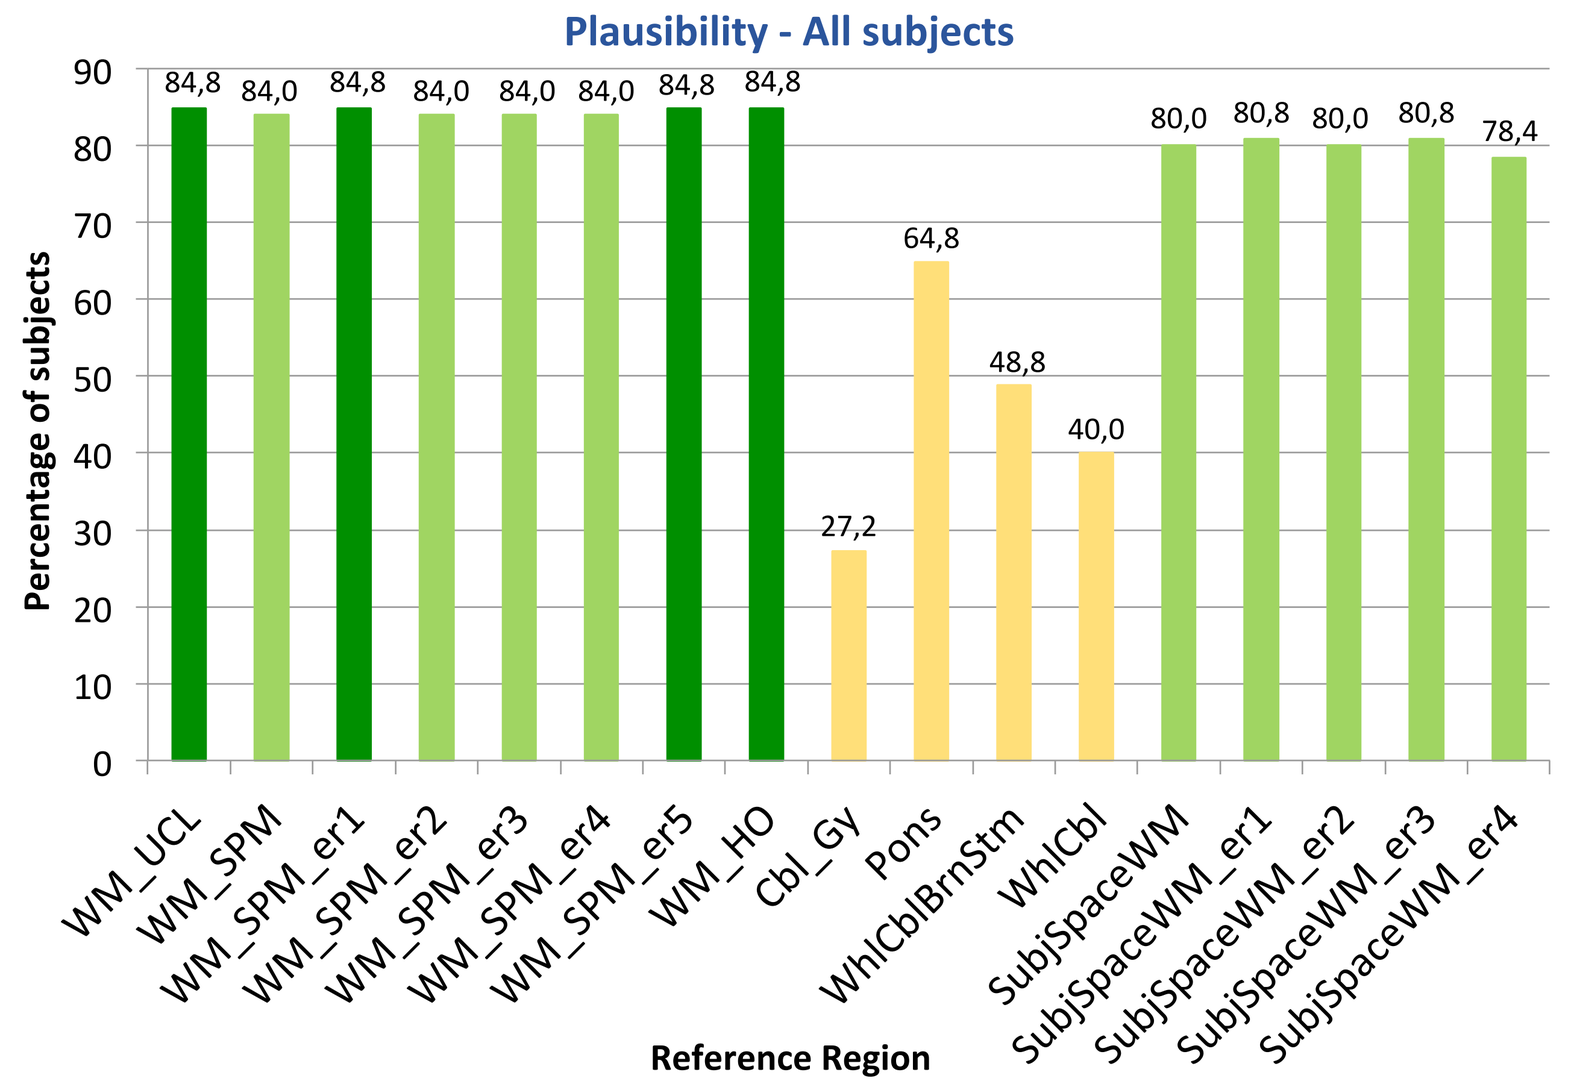
Plausibility - All subjects
Percentage of subjects
84,8 84,0 84,8 84,0 84,0 84,0 84,8 84,8 27,2 64,8 48,8 40,0 80,0 80,8 80,0 80,8 78,4
Reference Region

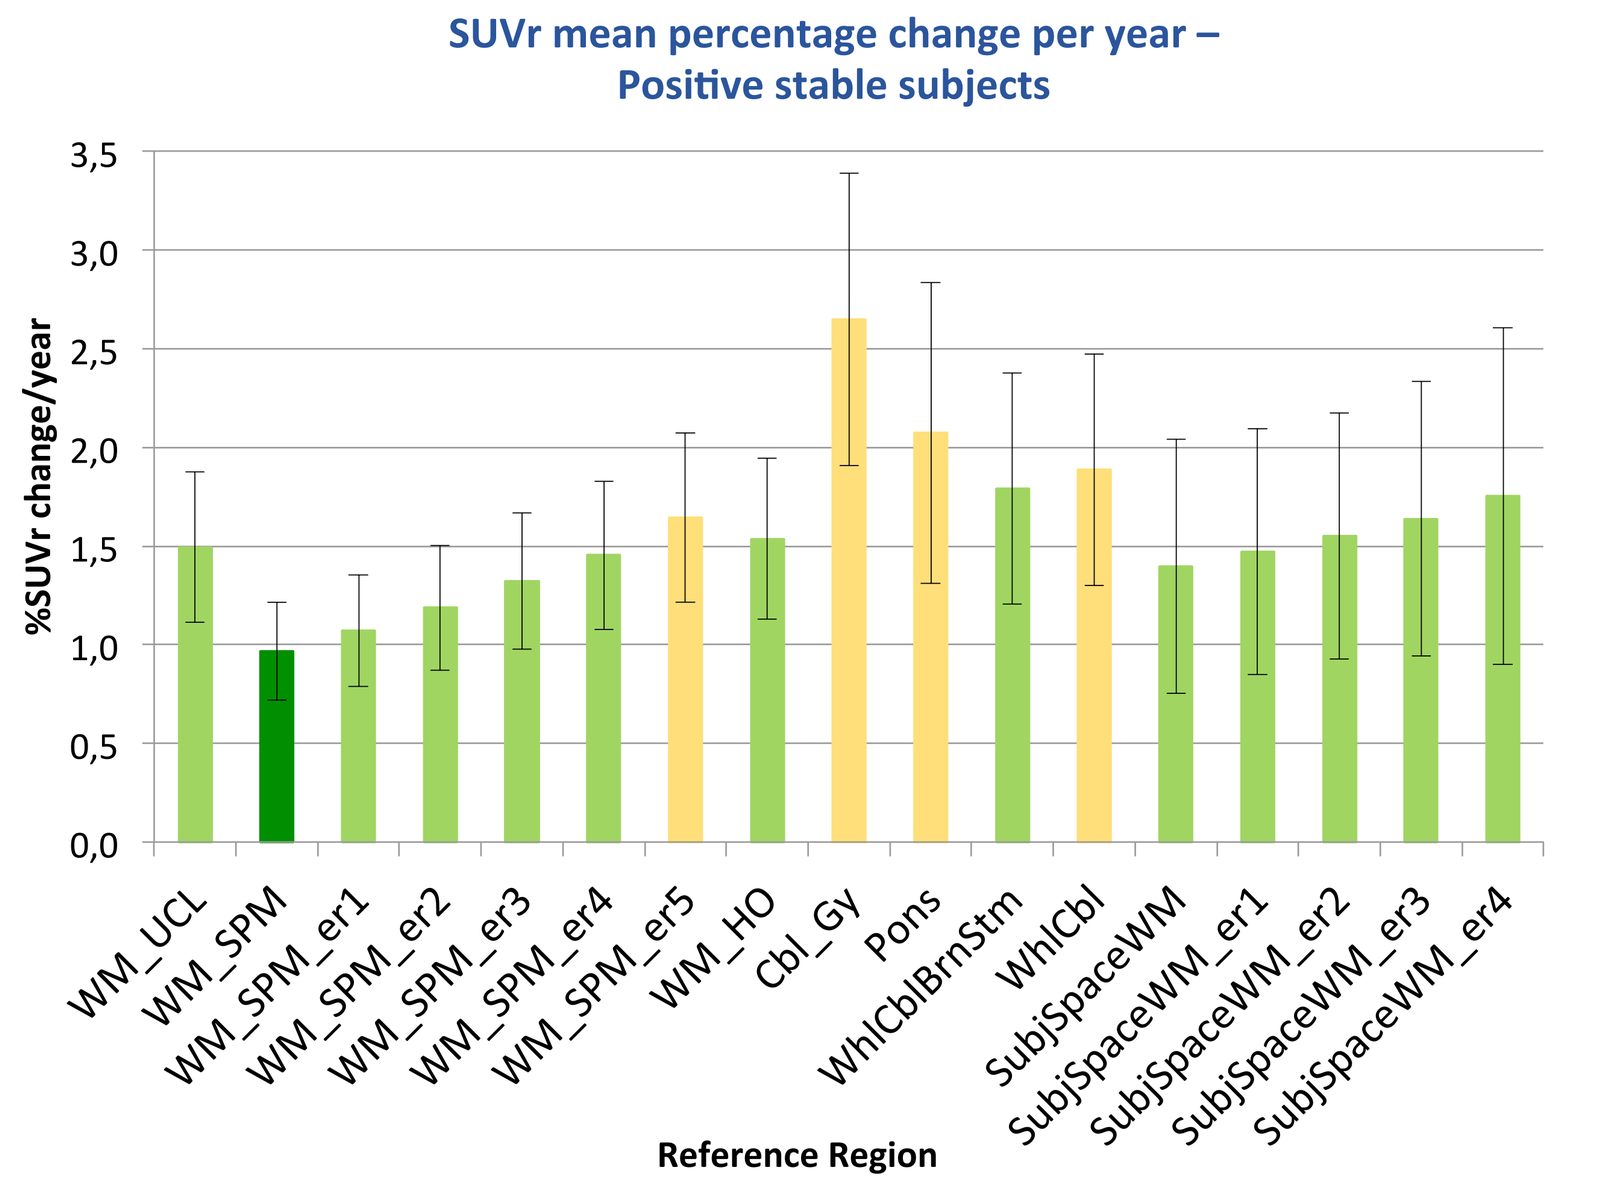
SUVr mean percentage change per year –
Positive stable subjects
%SUVr change/year
Reference Region

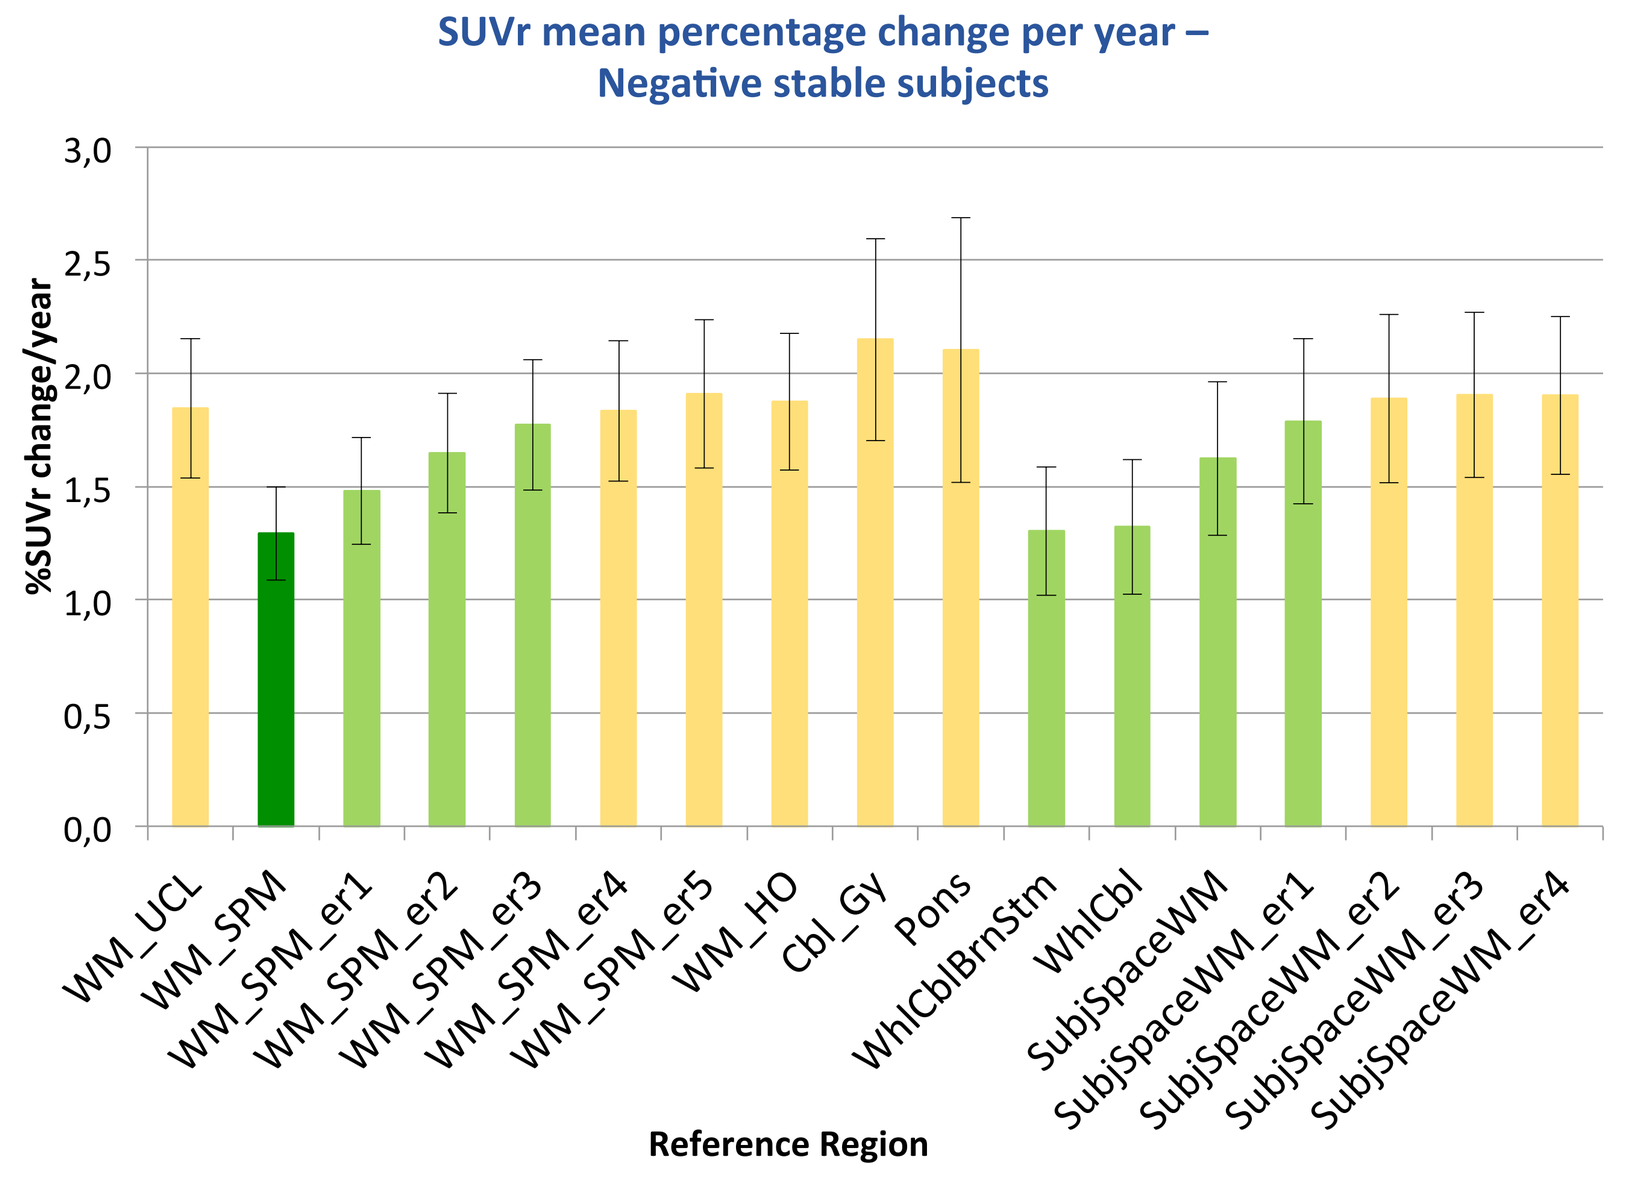
SUVr mean percentage change per year –
Negative stable subjects
%SUVr change/year
Reference Region

Longitudinal pipeline

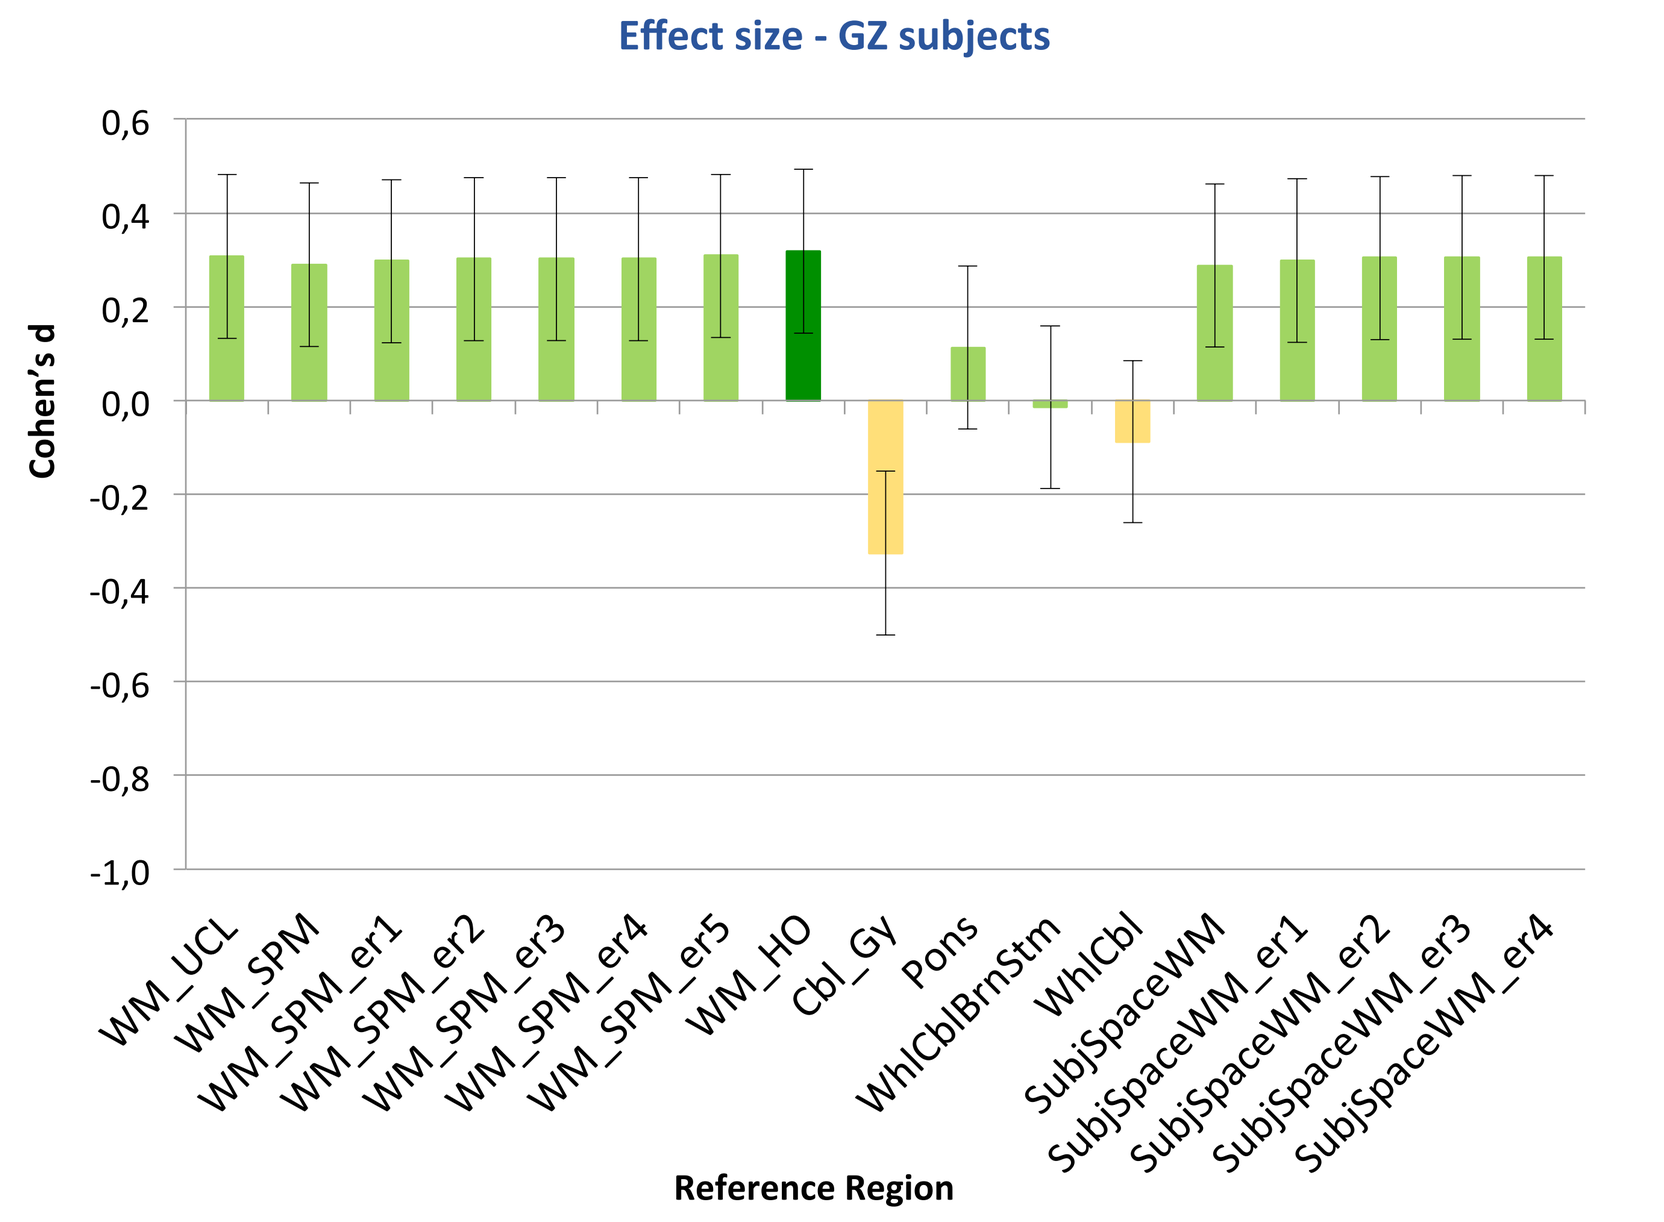
Effect size - GZ subjects
Cohen's d
Reference Region

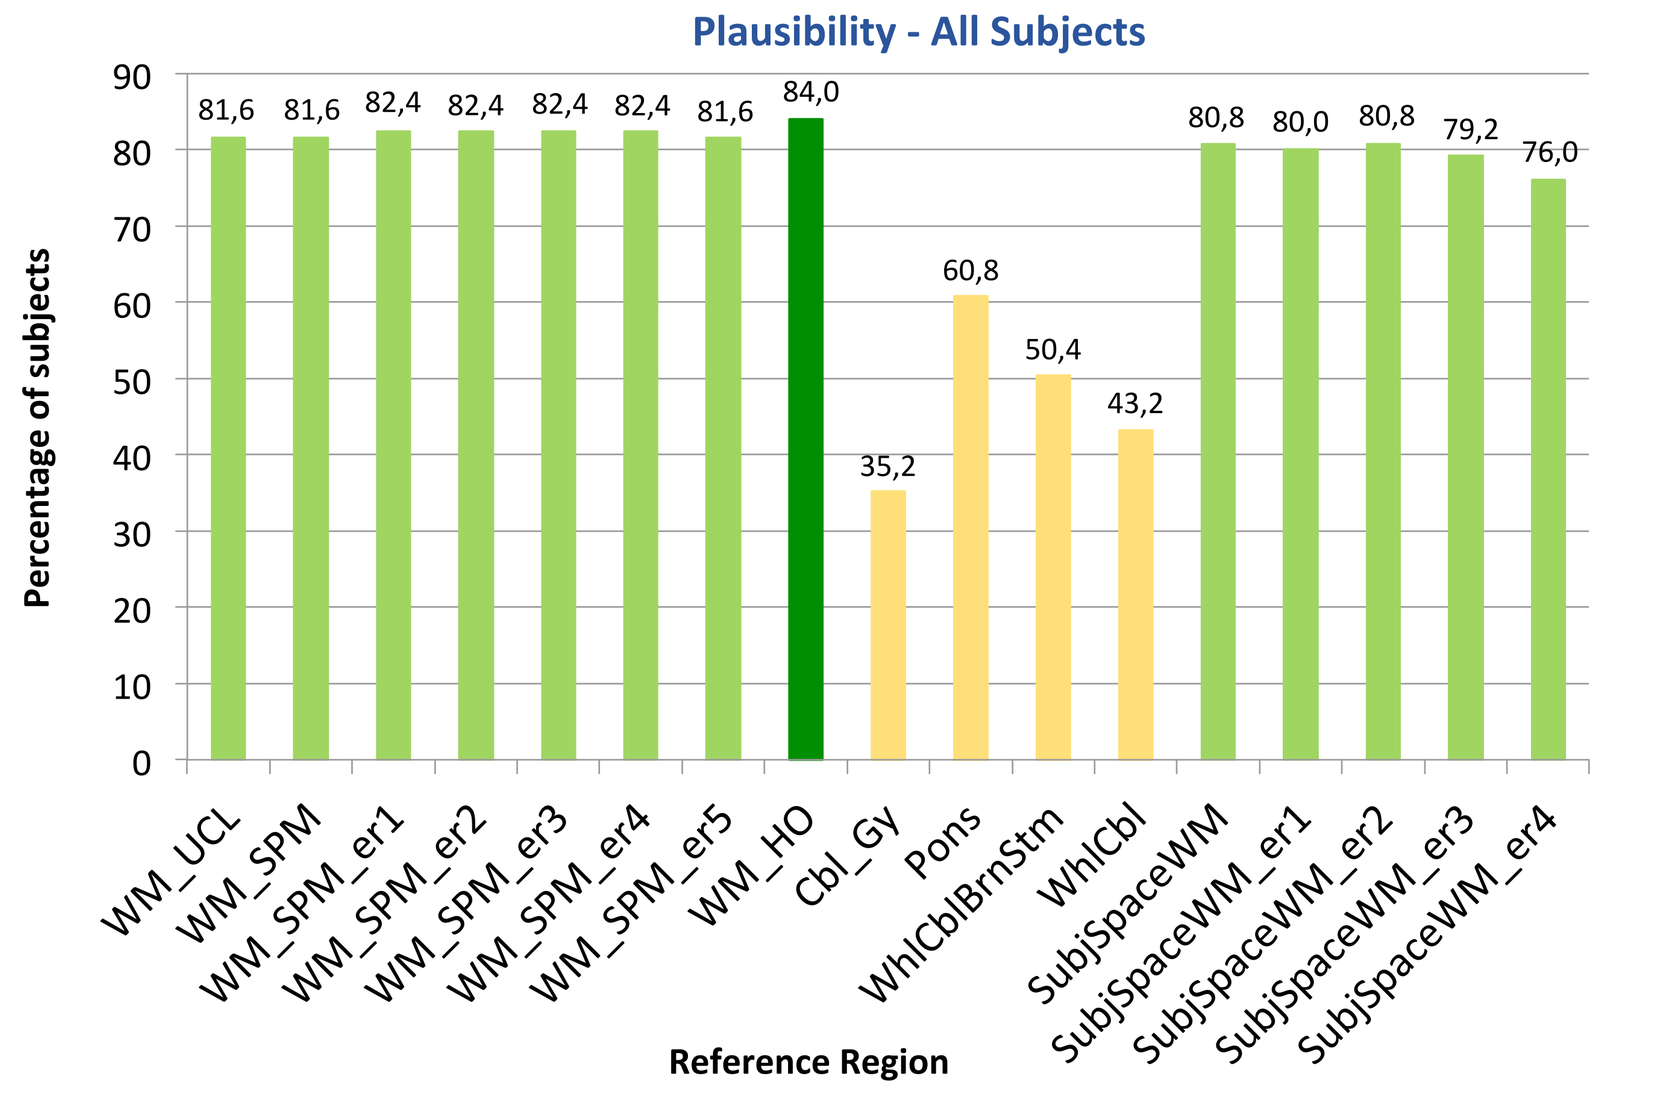
Plausibility - All Subjects
Percentage of subjects
81,6 81,6 82,4 82,4 82,4 82,4 81,6 84,0 35,2 60,8 50,4 43,2 80,8 80,0 80,8 79,2 76,0
Reference Region

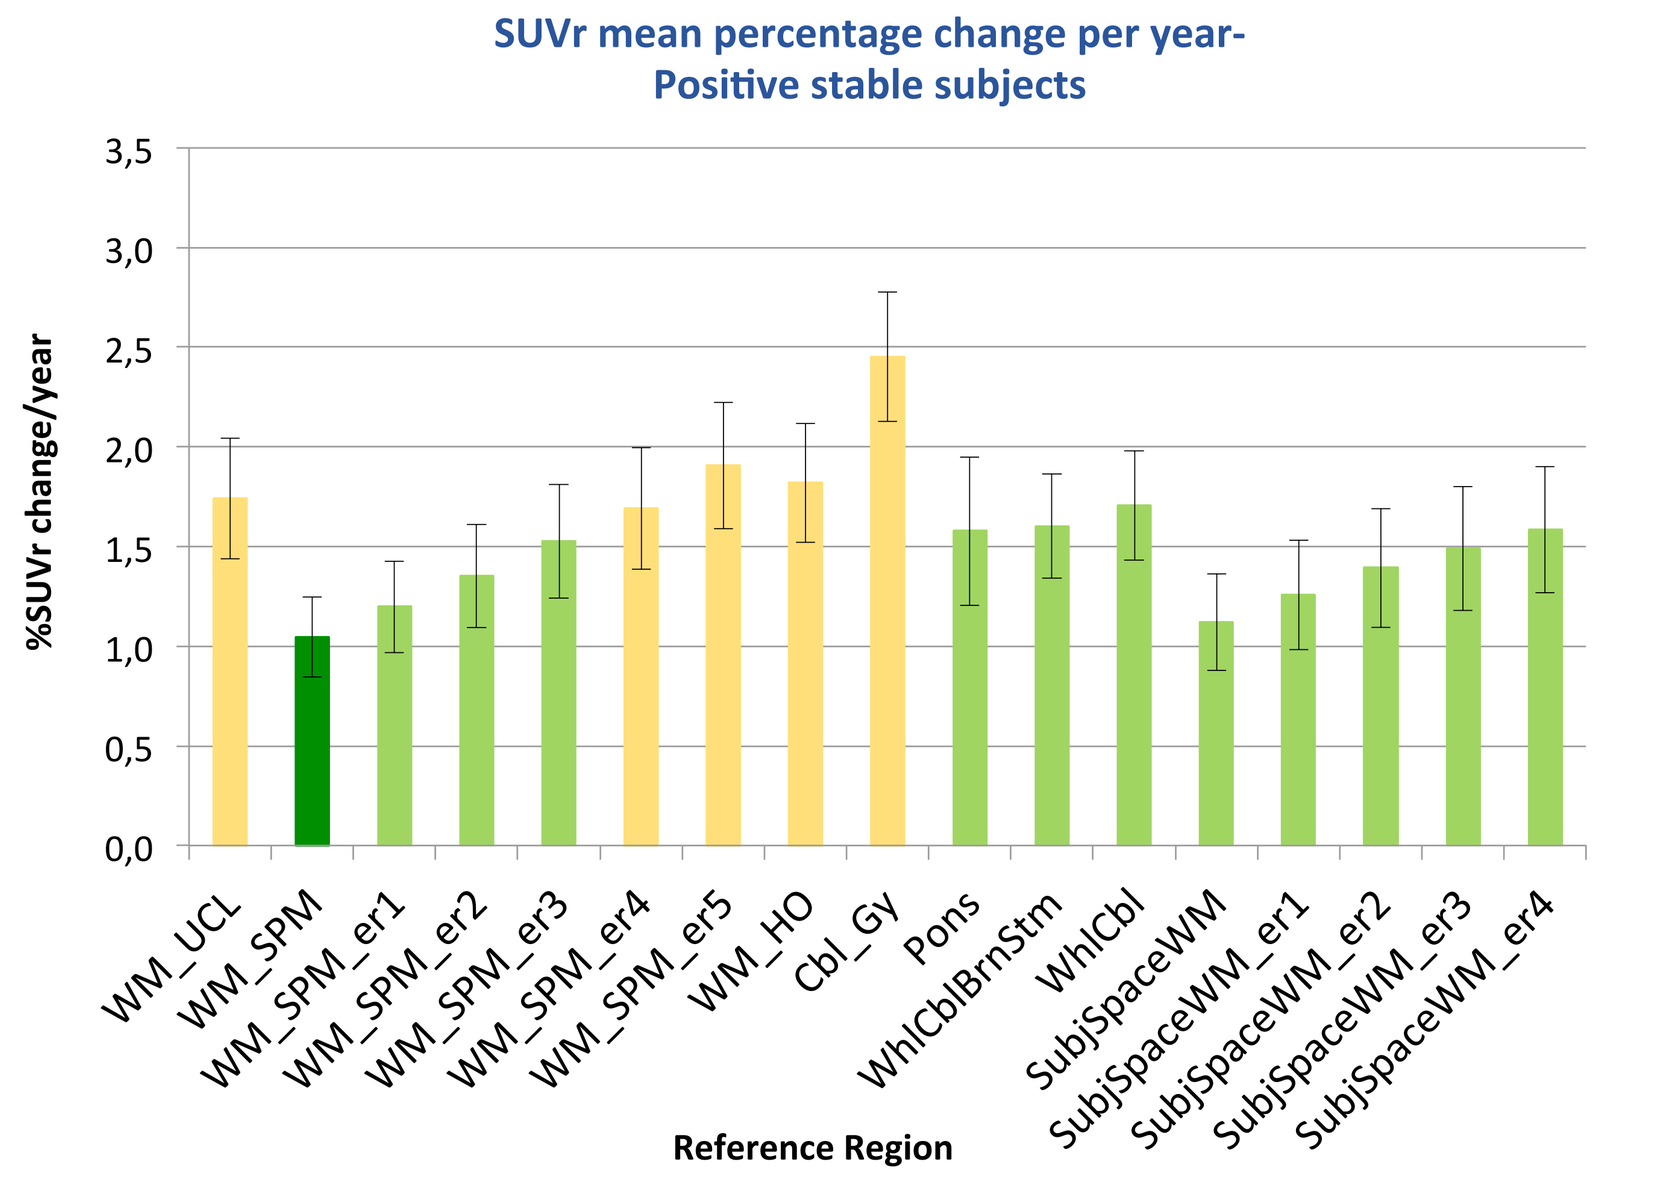
SUVr mean percentage change per year-
Positive stable subjects
%SUVr change/year
Reference Region

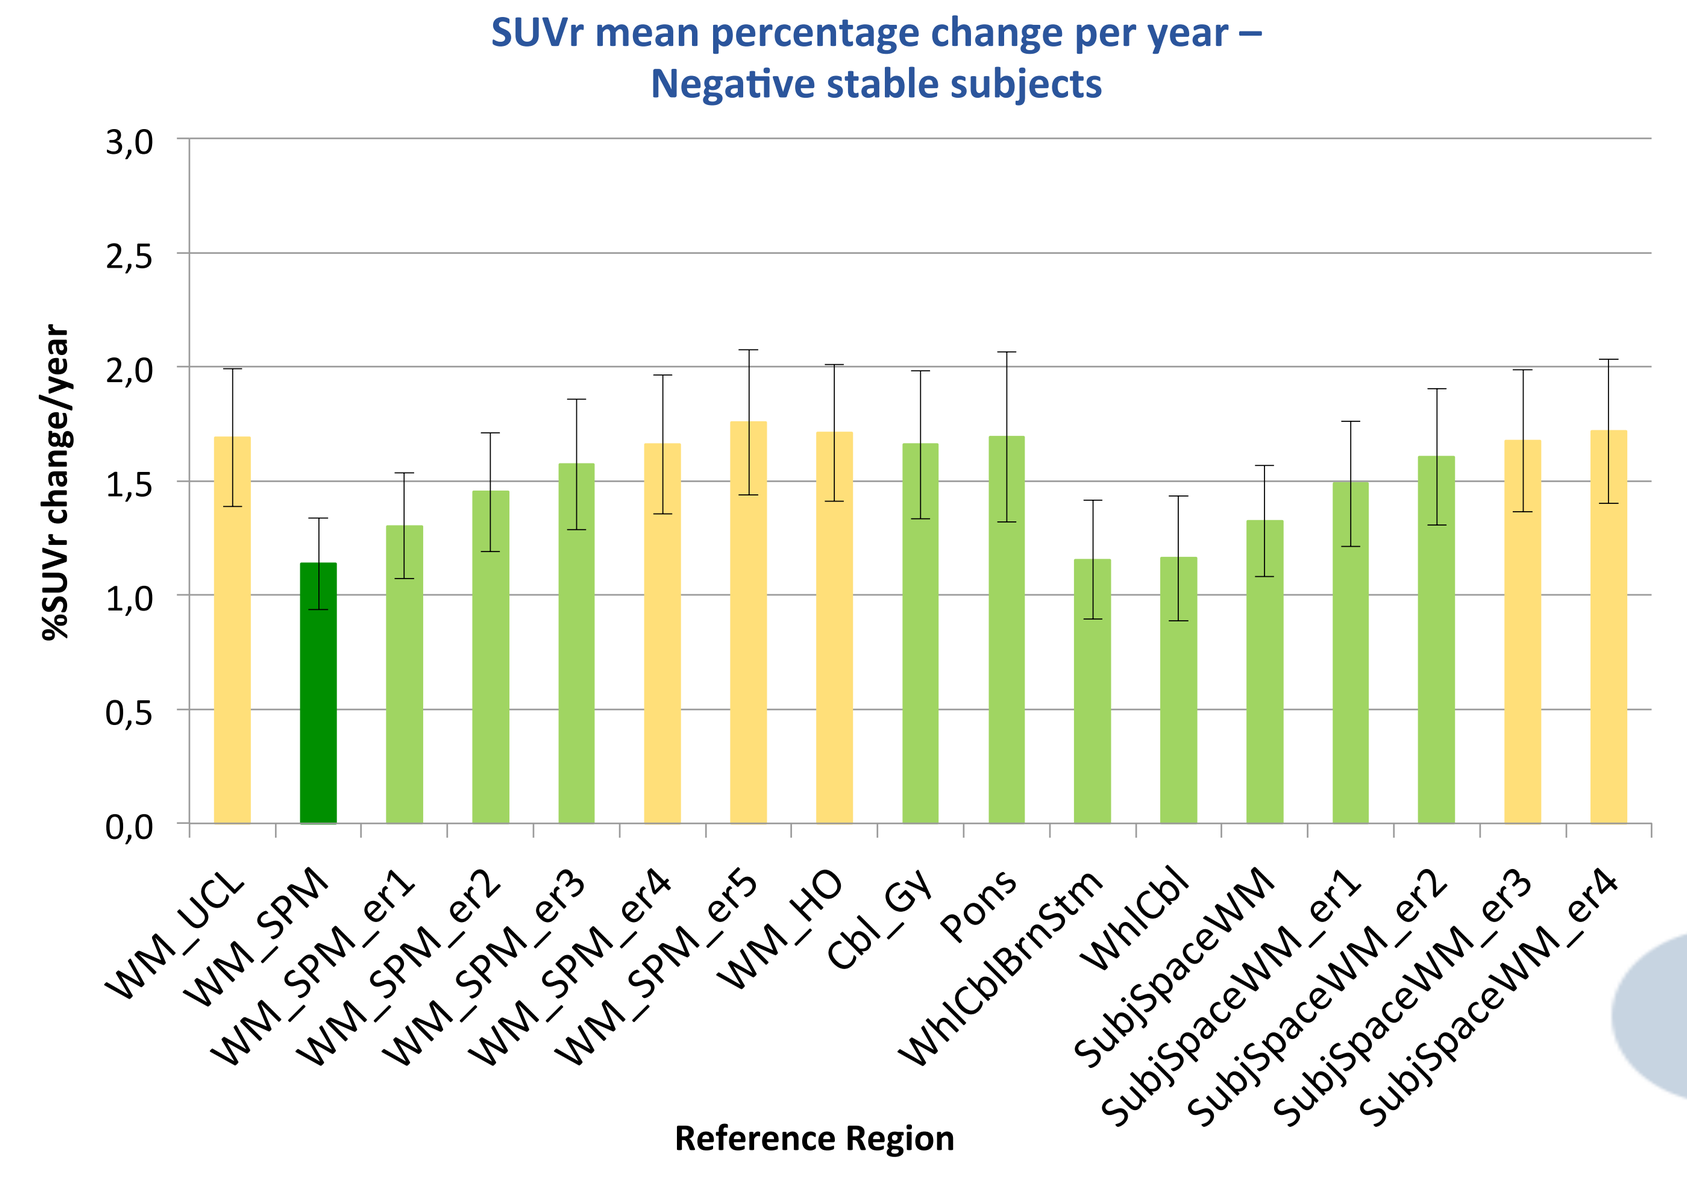
SUVr mean percentage change per year –
Negative stable subjects
%SUVr change/year
Reference Region

Discussion

Our results are in agreement with Landau *et al.* [4] that found that subcortical WM resulted more accurate to measure changes of Aβ with [^{18}F]Florbetapir. On the other hand, Bullich *et al.* [5] found that cerebellar RRs were optimal for detecting Aβ changes with [^{18}F]Florbetaben in MCI subjects.
A possible drawback for sWM as RR is that WM is susceptible of display pathological alterations of vascular origin in older individuals and MCI/AD patients that could affect tracer binding. Finally, the pipelines have been implemented on SPM, which might have favored the SPM-based RR.

Conclusions

- ✓ **sWM perform better than standard Centiloid RRs for longitudinal quantification**
- ✓ **Atlas-based sWM, which allow an easier automation, show equivalent performance as subject-specific RR**
- ✓ **The agreement between results with both pipelines suggest that our findings are generalizable to other pipeline implementations**

Acknowledgements

Authors would like to acknowledge Dr. Christopher C. Rowe on behalf of AIBL for sharing [^{18}F]Flutemetamol PET scans with the AMYPAD consortium.

References: [1] Klunk W. et al., Alzheimer's & Dementia (2014), [2] Ellis K.A. et al., International Psychogeriatrics (2009), [3] Jack C. Jr. et al., Neurology (2013), [4] Landau S.M. et al., J Nucl Med (2015), [5] Bullich S. et al. J Nucl Med (2017)

Contact: gsalvado@barcelonabeta.org

The project leading to this application has received funding from the Innovative Medicines Initiative 2 Joint Undertaking under grant agreement No 115952. This Joint Undertaking receives the support from the European Union's Horizon 2020 research and innovation programme and EFPIA.